Specifying Models with Time-Dependent Pharmacokinetic Parameters in NONMEM

A Brochot, A Dunne, I Poggesi, A Vermeulen

Advanced Modeling & Simulation, Janssen Research & Development

Beerse, Belgium

Introduction

Time-dependent parameters are sometimes required to describe the pharmacokinetics of a drug. Such models can be implemented in NONMEM in a number of different ways [1].

Two of them use the PREDPP library and were considered in this work :

- using an analytical solution
- using a numerical solution to the differential equations

The differences between these methods were investigated using an example of time-varying absorption previously presented [2,3].

Methods

Model

A one-compartment pharmacokinetic model with a time-dependent first order oral absorption was applied.

The time dependency of the absorption rate constant was described using a sigmoidal Emax model:

$$KA(t) = KA_{final} \cdot \frac{t^{\gamma}}{t_{50}^{\gamma} + t^{\gamma}}$$

where KA_{final} is the asymptotic value of KA, t_{50} is the time at which KA reaches 50% of its final values, and γ is a shape factor that modulates the onset of KA_{final}.

The model was parameterized using apparent clearance and volume. No inter- nor intra-subject variability was introduced in the model to facilitate relevant comparisons to be made.

Dataset

Two datasets were simulated, one using the analytical solution and the other using the numerical solution to the differential equations. Each dataset contained 3 individuals, receiving the same dose level of 100mg but having a different sampling scheme (from very rich to sparse) as illustrated in Table 1.

Table 1: Overview of the sampling schemes in the dataset

Sampling Time (hours)		
ID1	**ID2**	**ID3**
0.25	-	-
0.5	0.5	-
0.75	-	-
1	1	-
1.25	-	-
1.5	-	-
2	2	2
2.5	-	-
3	3	3
4	4	-
5	-	-
6	6	6
8	8	8
10	-	-
12	12	12
16	-	-
24	24	24

Implementation

Simulations were performed in NONMEM (v.7) [1] using two different control streams. The first control stream used the analytical solution in ADVAN2, whilst the second control stream used a numerical solution through ADVAN6. The NONMEM control streams are shown in Figure 1 and Figure 2, respectively.

Figure 1: Control stream using the analytical solution

```
$PROBLEM RUN #001
$INPUT C ID AMT MDV EVID TIME DV CMT DOSE
$DATA sim_dataset.csv IGNORE='C'
$SUBROUTINES  ADVAN2 TRANS2

$PK
  TVCL = THETA(1)
  CL   = TVCL
  TVV  = THETA(2)
  V    = TVV
  TVKA = THETA(3)
  TT50 = THETA(4)
  GAM  = THETA(5)

  KA   = TVKA*TIME**GAM/(TT50**GAM+TIME**GAM)
```

Figure 2: Control stream using the numerical solution to the differential equations

```
$PROBLEM RUN #001
$INPUT C ID AMT MDV EVID TIME DV CMT DOSE
$DATA sim_dataset.csv IGNORE='C'
$SUBROUTINES  ADVAN6 TOL=5
$MODEL
   COMP=(DEPOT,DEFDOSE)
   COMP=(CENTRAL,DEFOBS)

$PK
  TVCL = THETA(1)
  CL   = TVCL
  TVV  = THETA(2)
  V    = TVV
  TVKA = THETA(3)
  TT50 = THETA(4)
  GAM  = THETA(5)
  KA   = TVKA
  K20  = CL/V

$DES
   DADT(1) = -KA*T**GAM/(TT50**GAM+T**GAM)*A(1)
   DADT(2) =  KA*T**GAM/(TT50**GAM+T**GAM)*A(1)  - K20*A(2)
```

Results

The IPRED values up to Tmax simulated with the two control streams are given in Table 2 and Table 3.

Table 2: Results from simulation using control stream in Figure 1

ID	TIME	IPRED
1	0.25	26.8
1	**0.5**	**218**
1	0.75	374
1	**1**	**467**
1	1.25	516
1	1.5	537
1	**2**	**532**
2	**0.5**	**333**
2	**1**	**503**
2	**2**	**521**
3	**2**	**517**

Table 3: Results from simulation using control stream in Figure 2

ID	TIME	IPRED
1	0.25	4.81
1	**0.5**	**124**
1	0.75	307
1	**1**	**428**
1	1.25	497
1	1.5	530
1	**2**	**537**
2	**0.5**	**124**
2	**1**	**428**
2	**2**	**537**
3	**2**	**537**

The most notable feature about these tables of results is that in Table 2, the three subjects have different IPRED values at the common time points **0.5**, **1** and **3** hours, despite there being no inter- nor intra-subject variability. On the contrary, Table 3 does not show any difference between subjects as would be expected with this model.

Because the analytical solution implemented by ADVAN2 treats the KA values as being fixed between observation times, the model for KA being simulated using the code in Figure 1 can be described by the step functions illustrated in Figure 3. It can be appreciated that this model differs from subject to subject if the sampling scheme differs between individuals.

Figure 3: The models for the absorption rate constant (KA) as simulated by the control stream in Figure 1 (red line) and by the control stream in Figure 2 (blue line) (top: ID1, middle: ID2, bottom: ID3)

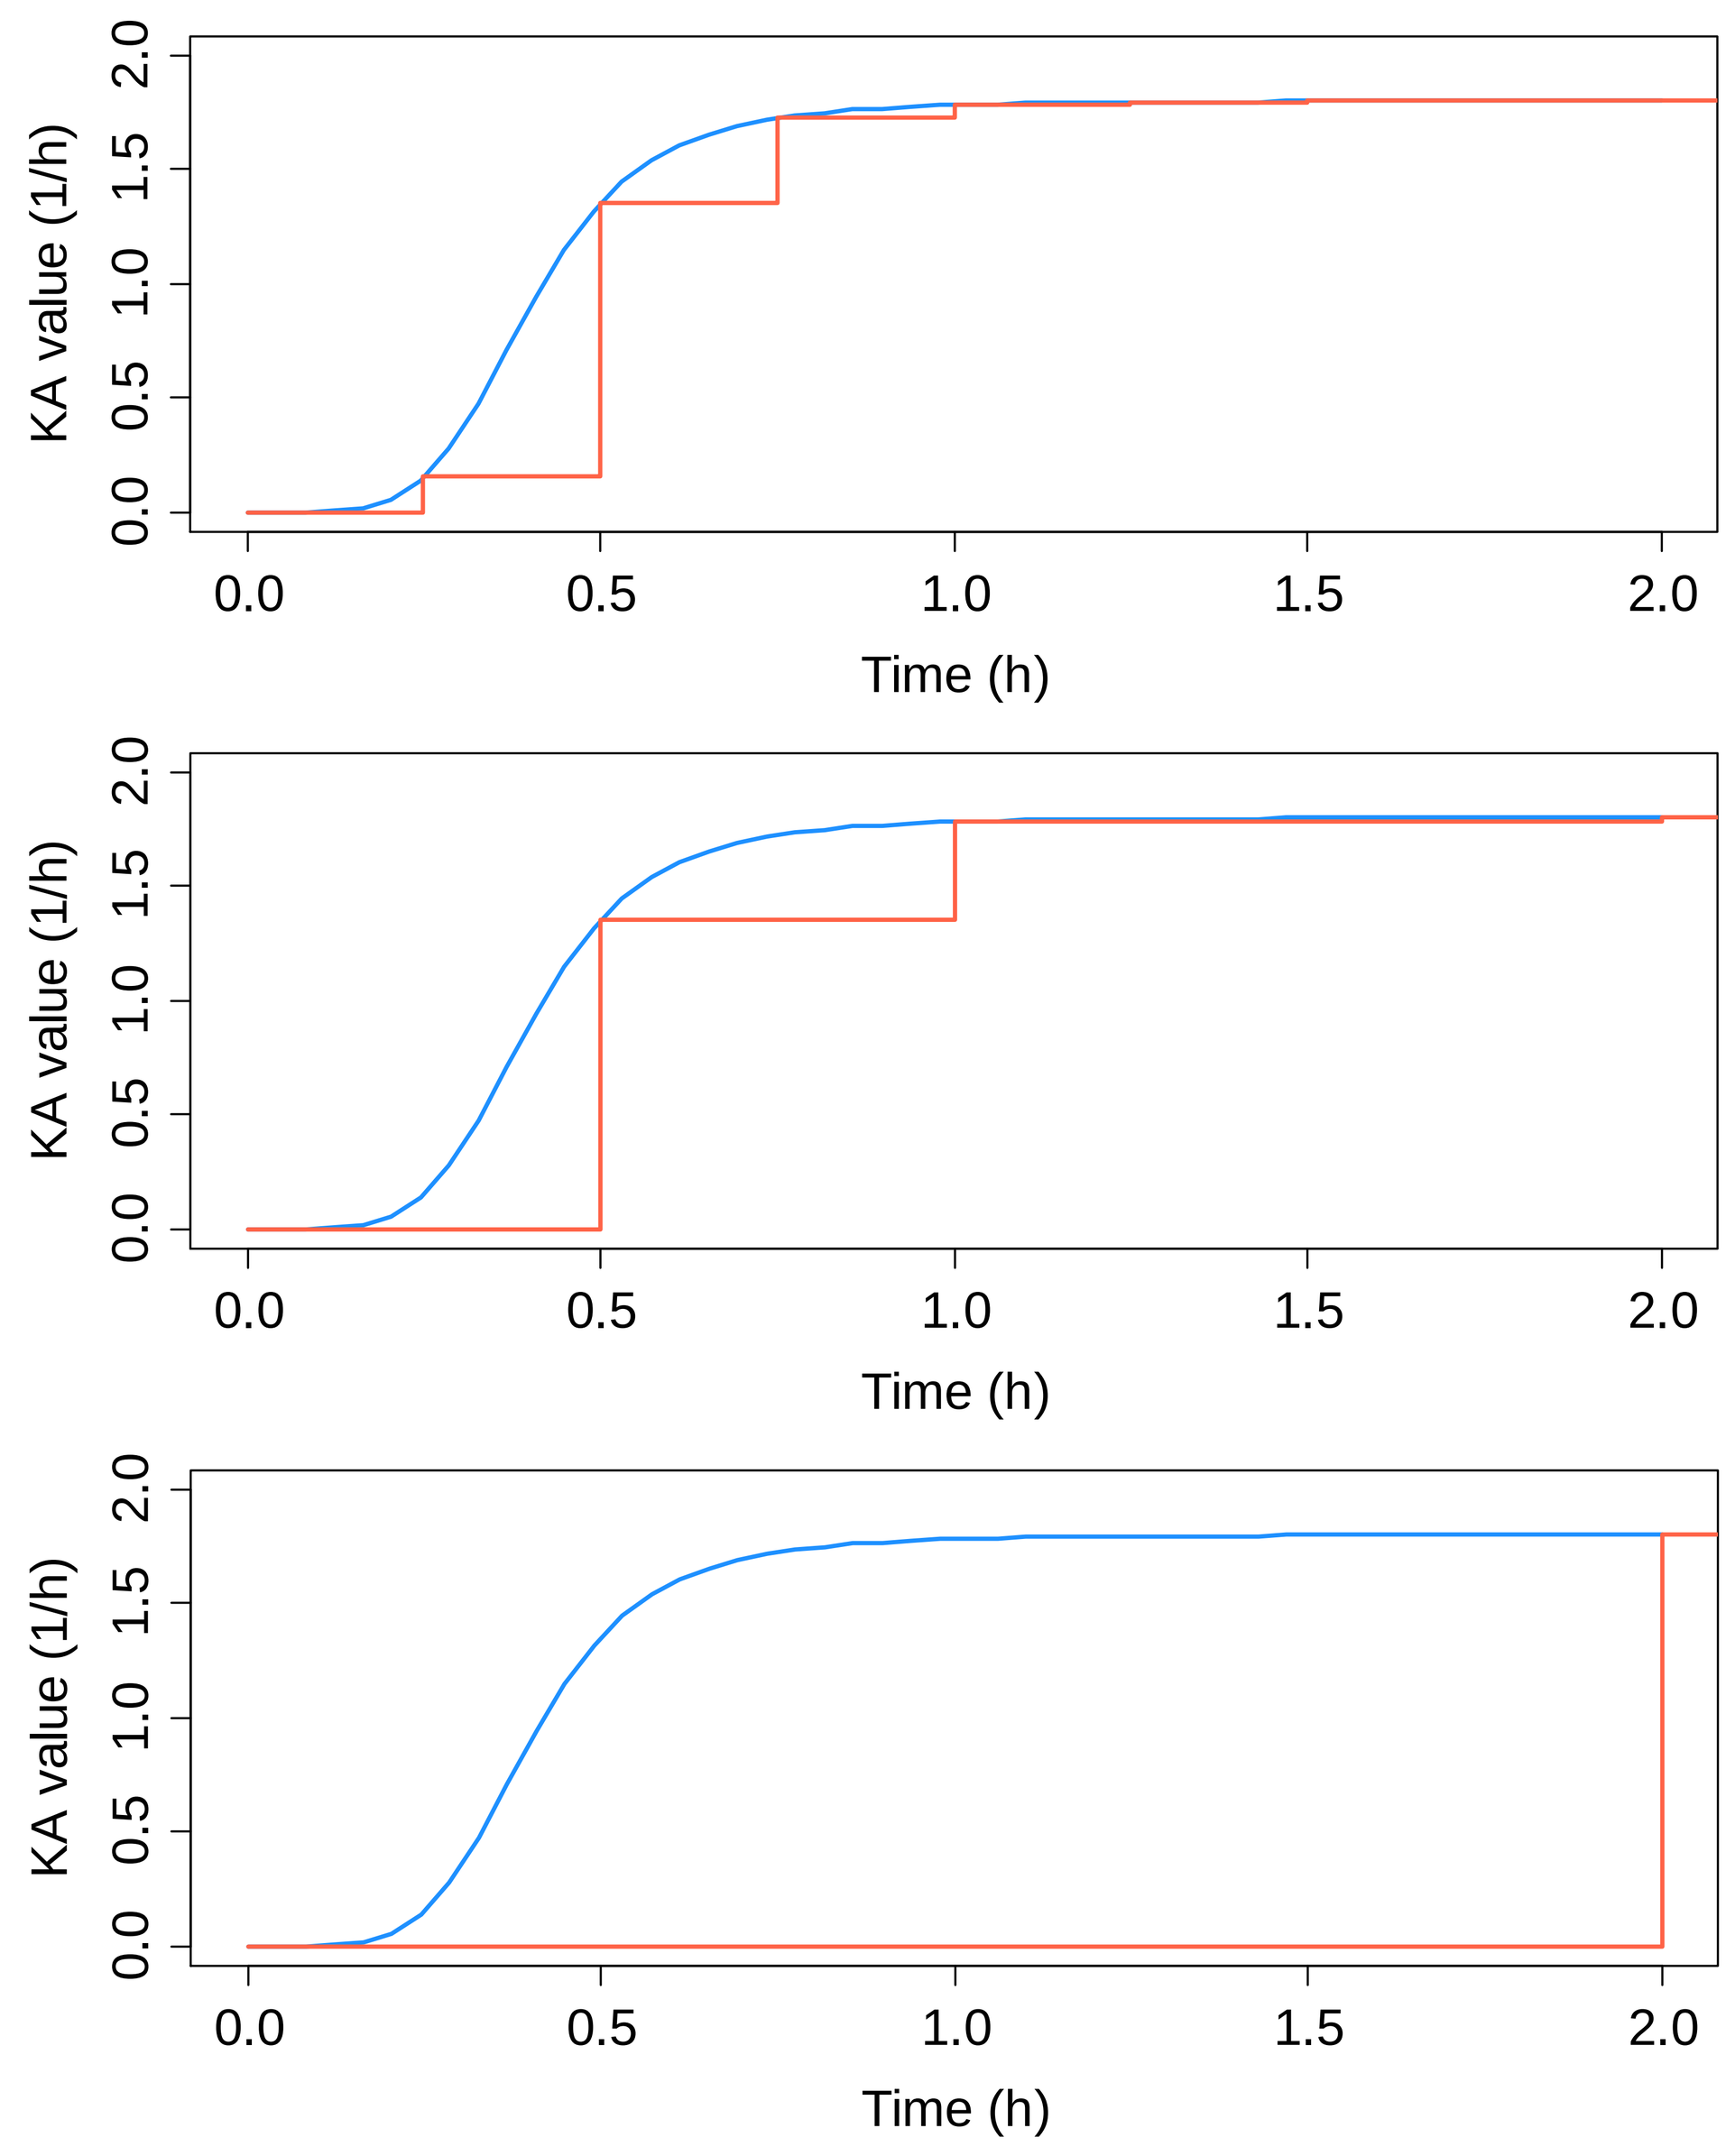

KA value (1/h)
Time (h)

Figure 4 illustrates the deviation of the IPRED values when using the control stream from Figure 1 versus that from Figure 2 which gives the exact solution. This figure proves that a richer sampling scheme as ID1 resullts in a higher precision.

Figure 4: IPRED simulated using control stream in Figure 1 versus IPRED simulated using control stream in Figure 2 for the 3 subjects up to Tmax. Blue line is the identity line

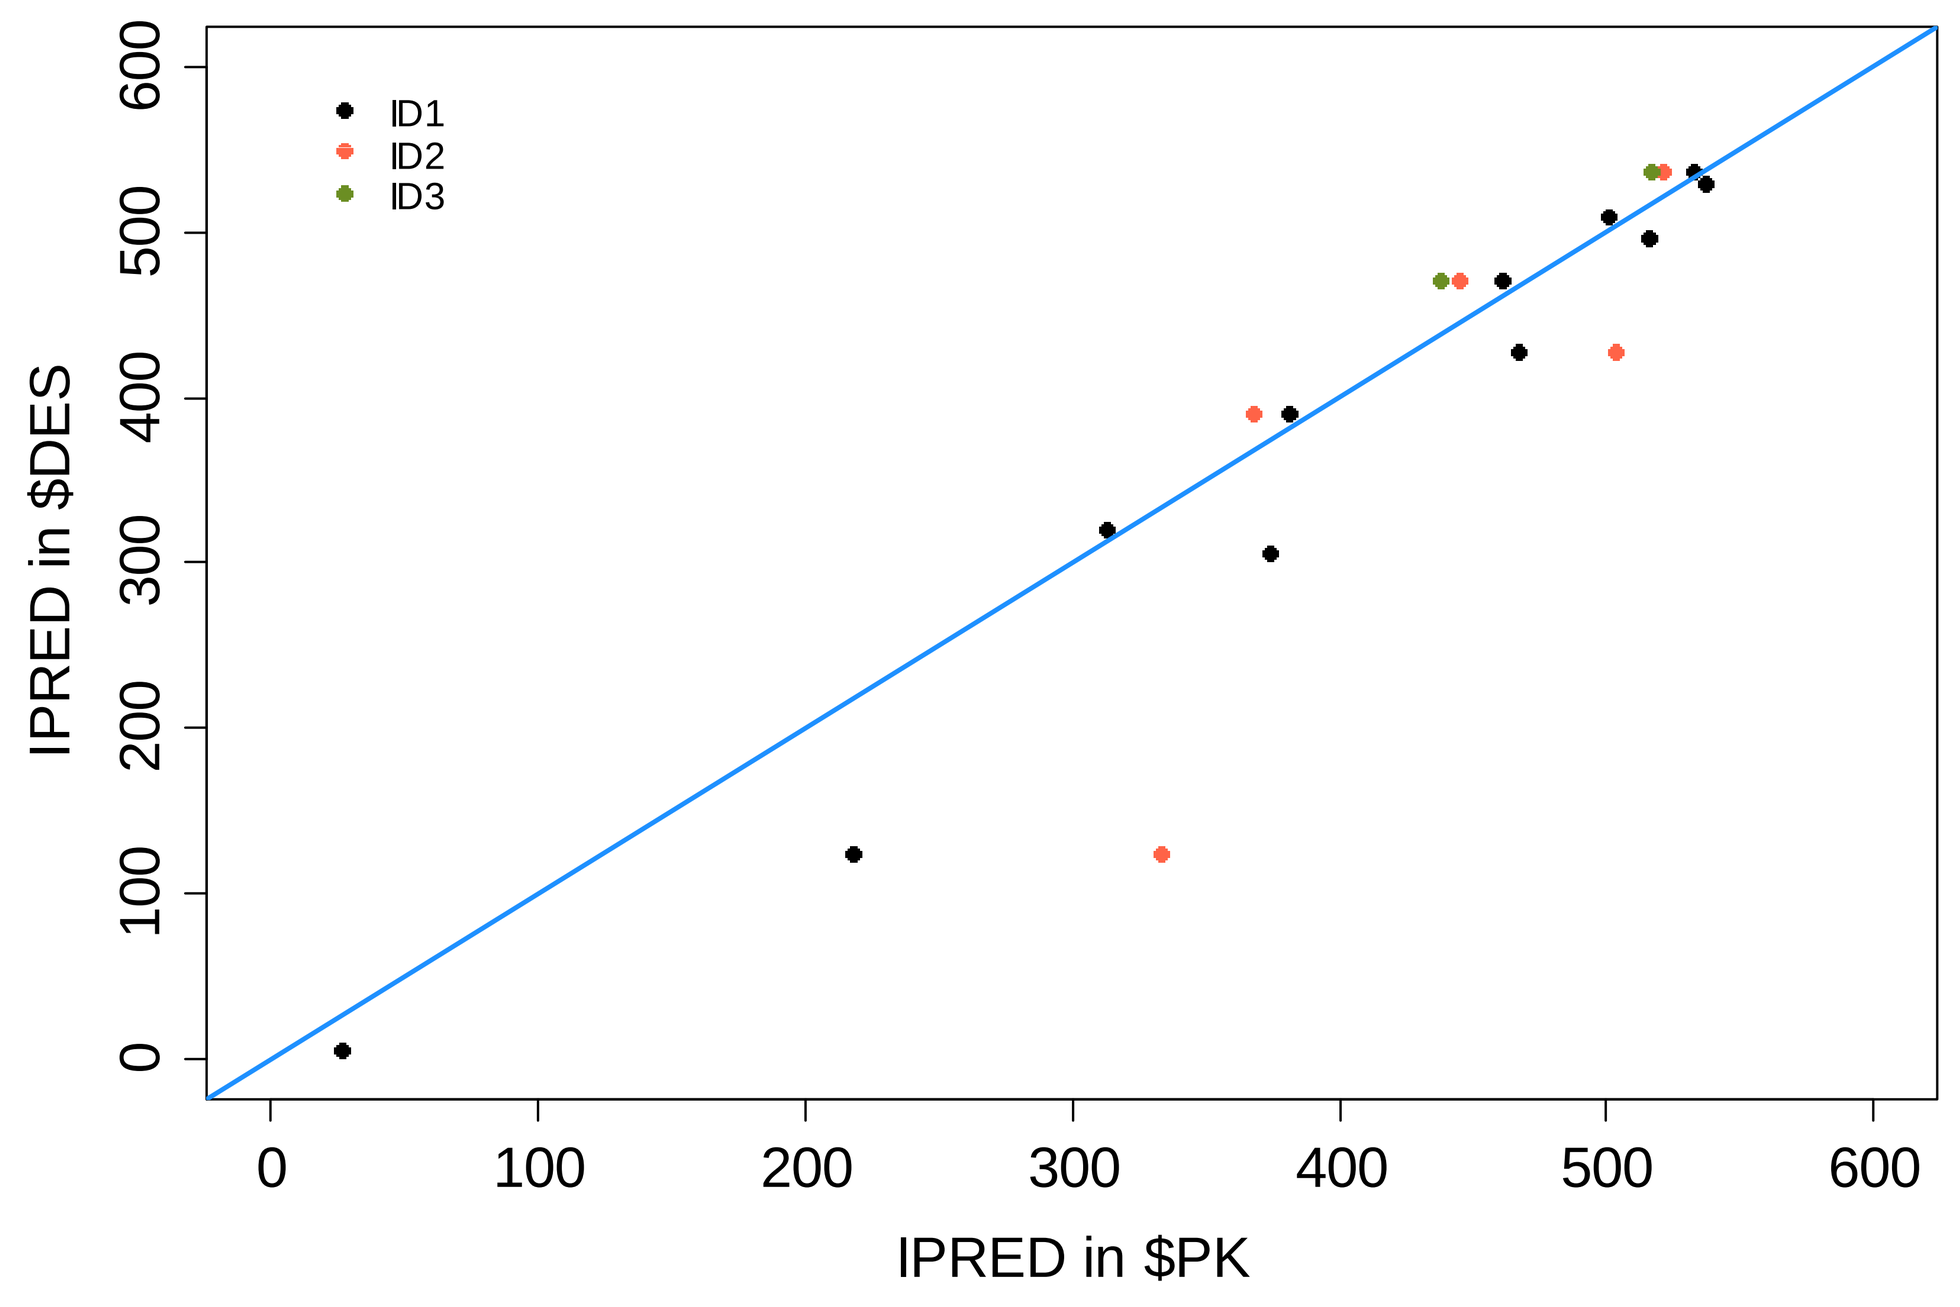

ID1
ID2
ID3
IPRED in $DES
IPRED in $PK

Conclusion

It is clear from Figure 3 and Figure 4 that the control stream using the analytical solution (ADVAN2) does not use the model as described in Equation 1, and even uses a different model for each subject when the sampling scheme differs between individuals. The model obtained in this case is a step function approximation of the desired model.

On the other hand, the numerical differential equation solver in NONMEM uses the desired model but is more computer intensive.

If the analytical solution is used, the introduction of a sufficient number of time points is required to have an acceptable approximation of the model.

References

[1] Beal S et al. NONMEM User’s Guides. (1989-2009), Icon Development Solutions, Ellicott City, MD, USA.

[2] Iavarone et al., PAGE 19 (2010) Abstr 1856 [www.page-meeting.org/?abstract=1856].

[3] Petricoul O, Cosson V, Fuseau E, Marchand M. Population Models for drug absorption and enterohepatic recycling. Pharmacometrics: the science of quantitative pharmacology, 2007, Ette EI, Williams PJ (eds). John Wiley&Sons Inc.